Poster 29-P

Outcomes with finerenone in patients with chronic kidney disease and type 2 diabetes by baseline insulin resistance: A FIDELITY subgroup analysis

Thomas Ebert,[1] Stefan D. Anker,[2] Luis M. Ruilope,[3–5] Paola Fioretto,[6] Vivian Fonseca,[7] Guillermo Umpierrez,[8] Andreas L. Birkenfeld,[9,10] Robert Lawatscheck,[11] Charlie Scott,[12] Katja Rohwedder,[13] Peter Rossing,[14,15]
on behalf of the FIDELIO-DKD and FIGARO-DKD Investigators

[1]Medical Department III – Endocrinology, Nephrology, Rheumatology, University of Leipzig Medical Center, Leipzig, Germany; [2]Department of Cardiology (CVK) of German Heart Center Charité; Institute of Health Center for Regenerative Therapies (BCRT), German Centre for Cardiovascular Research (DZHK) partner Site Berlin, Charité Universitätsmedizin, Berlin, Germany; [3]Cardiorenal Translational Laboratory and Hypertension Unit, Institute of Research imas12, Madrid, Spain; [4]CIBER-CV, Hospital Universitario 12 de Octubre, Madrid, Spain; [5]Faculty of Sport Sciences, European University of Madrid, Madrid, Spain; [6]Department of Medicine, University of Padua, Italy; [7]Tulane University Health Sciences Center, New Orleans, LA, USA; [8]Division of Endocrinology, Emory University School of Medicine, Atlanta, Georgia, USA; [9]Department of Diabetology, Endocrinology and Nephrology, University Clinic, Tübingen, Germany; [10]Institute for Diabetes Research and Metabolic Diseases, Helmholtz Center Munich, University of Tübingen, Tübingen, Germany and German Center for Diabetes Research (DZD); [11]Clinical Research, Bayer AG, Berlin, Germany; [12]Data Science and Analytics, Bayer PLC, Reading, UK; [13]Medical Affairs, Bayer AG, Berlin, Germany; [14]Steno Diabetes Center Copenhagen, Herlev, Denmark; [15]Department of Clinical Medicine, University of Copenhagen, Copenhagen, Denmark

Professor Thomas Ebert • University of Leipzig Medical Center • thomas.ebert@medizin.uni-leipzig.de

1. Introduction

- Insulin resistance is associated with an increased risk of type 2 diabetes (T2D), cardiovascular (CV) disease, and chronic kidney disease (CKD)[1-3]
- Finerenone (a nonsteroidal mineralocorticoid receptor antagonist) improved cardiorenal outcomes in a broad population of patients with CKD and T2D in the FIDELITY prespecified pooled analysis[4] of the FIDELIO-DKD[5] and FIGARO-DKD[6] studies
- The aim of this post hoc analysis was to explore whether insulin resistance, measured by estimated glucose disposal rate (eGDR), is associated with increased risk of cardiorenal outcomes and if insulin resistance modifies the cardiorenal efficacy of finerenone

2. Methods

- This analysis combines individual patient-level data from the FIDELIO-DKD (NCT02540993) and FIGARO-DKD (NCT02545049) phase III clinical trials. The designs and results of these studies have been published previously[5,6]
- Study design, efficacy outcomes, and inclusion/exclusion criteria for FIDELITY are shown in **Figure 1**
- Insulin resistance was estimated using eGDR (an inverse marker of insulin resistance) and was calculated as follows: 21.158 + (–0.09 × waist circumference [cm]) + (–3.407 × presence of hypertension) + (–0.551 × HbA1c [%])
 - Lower eGDR is associated with greater insulin resistance and an increased risk of CV disease and progression to end-stage kidney disease versus higher eGDR (insulin sensitive)[1-3]
- Composite outcomes were analyzed by defined categorical subgroups: <median eGDR and ≥median eGDR at baseline

Figure 1. Study design and patient population

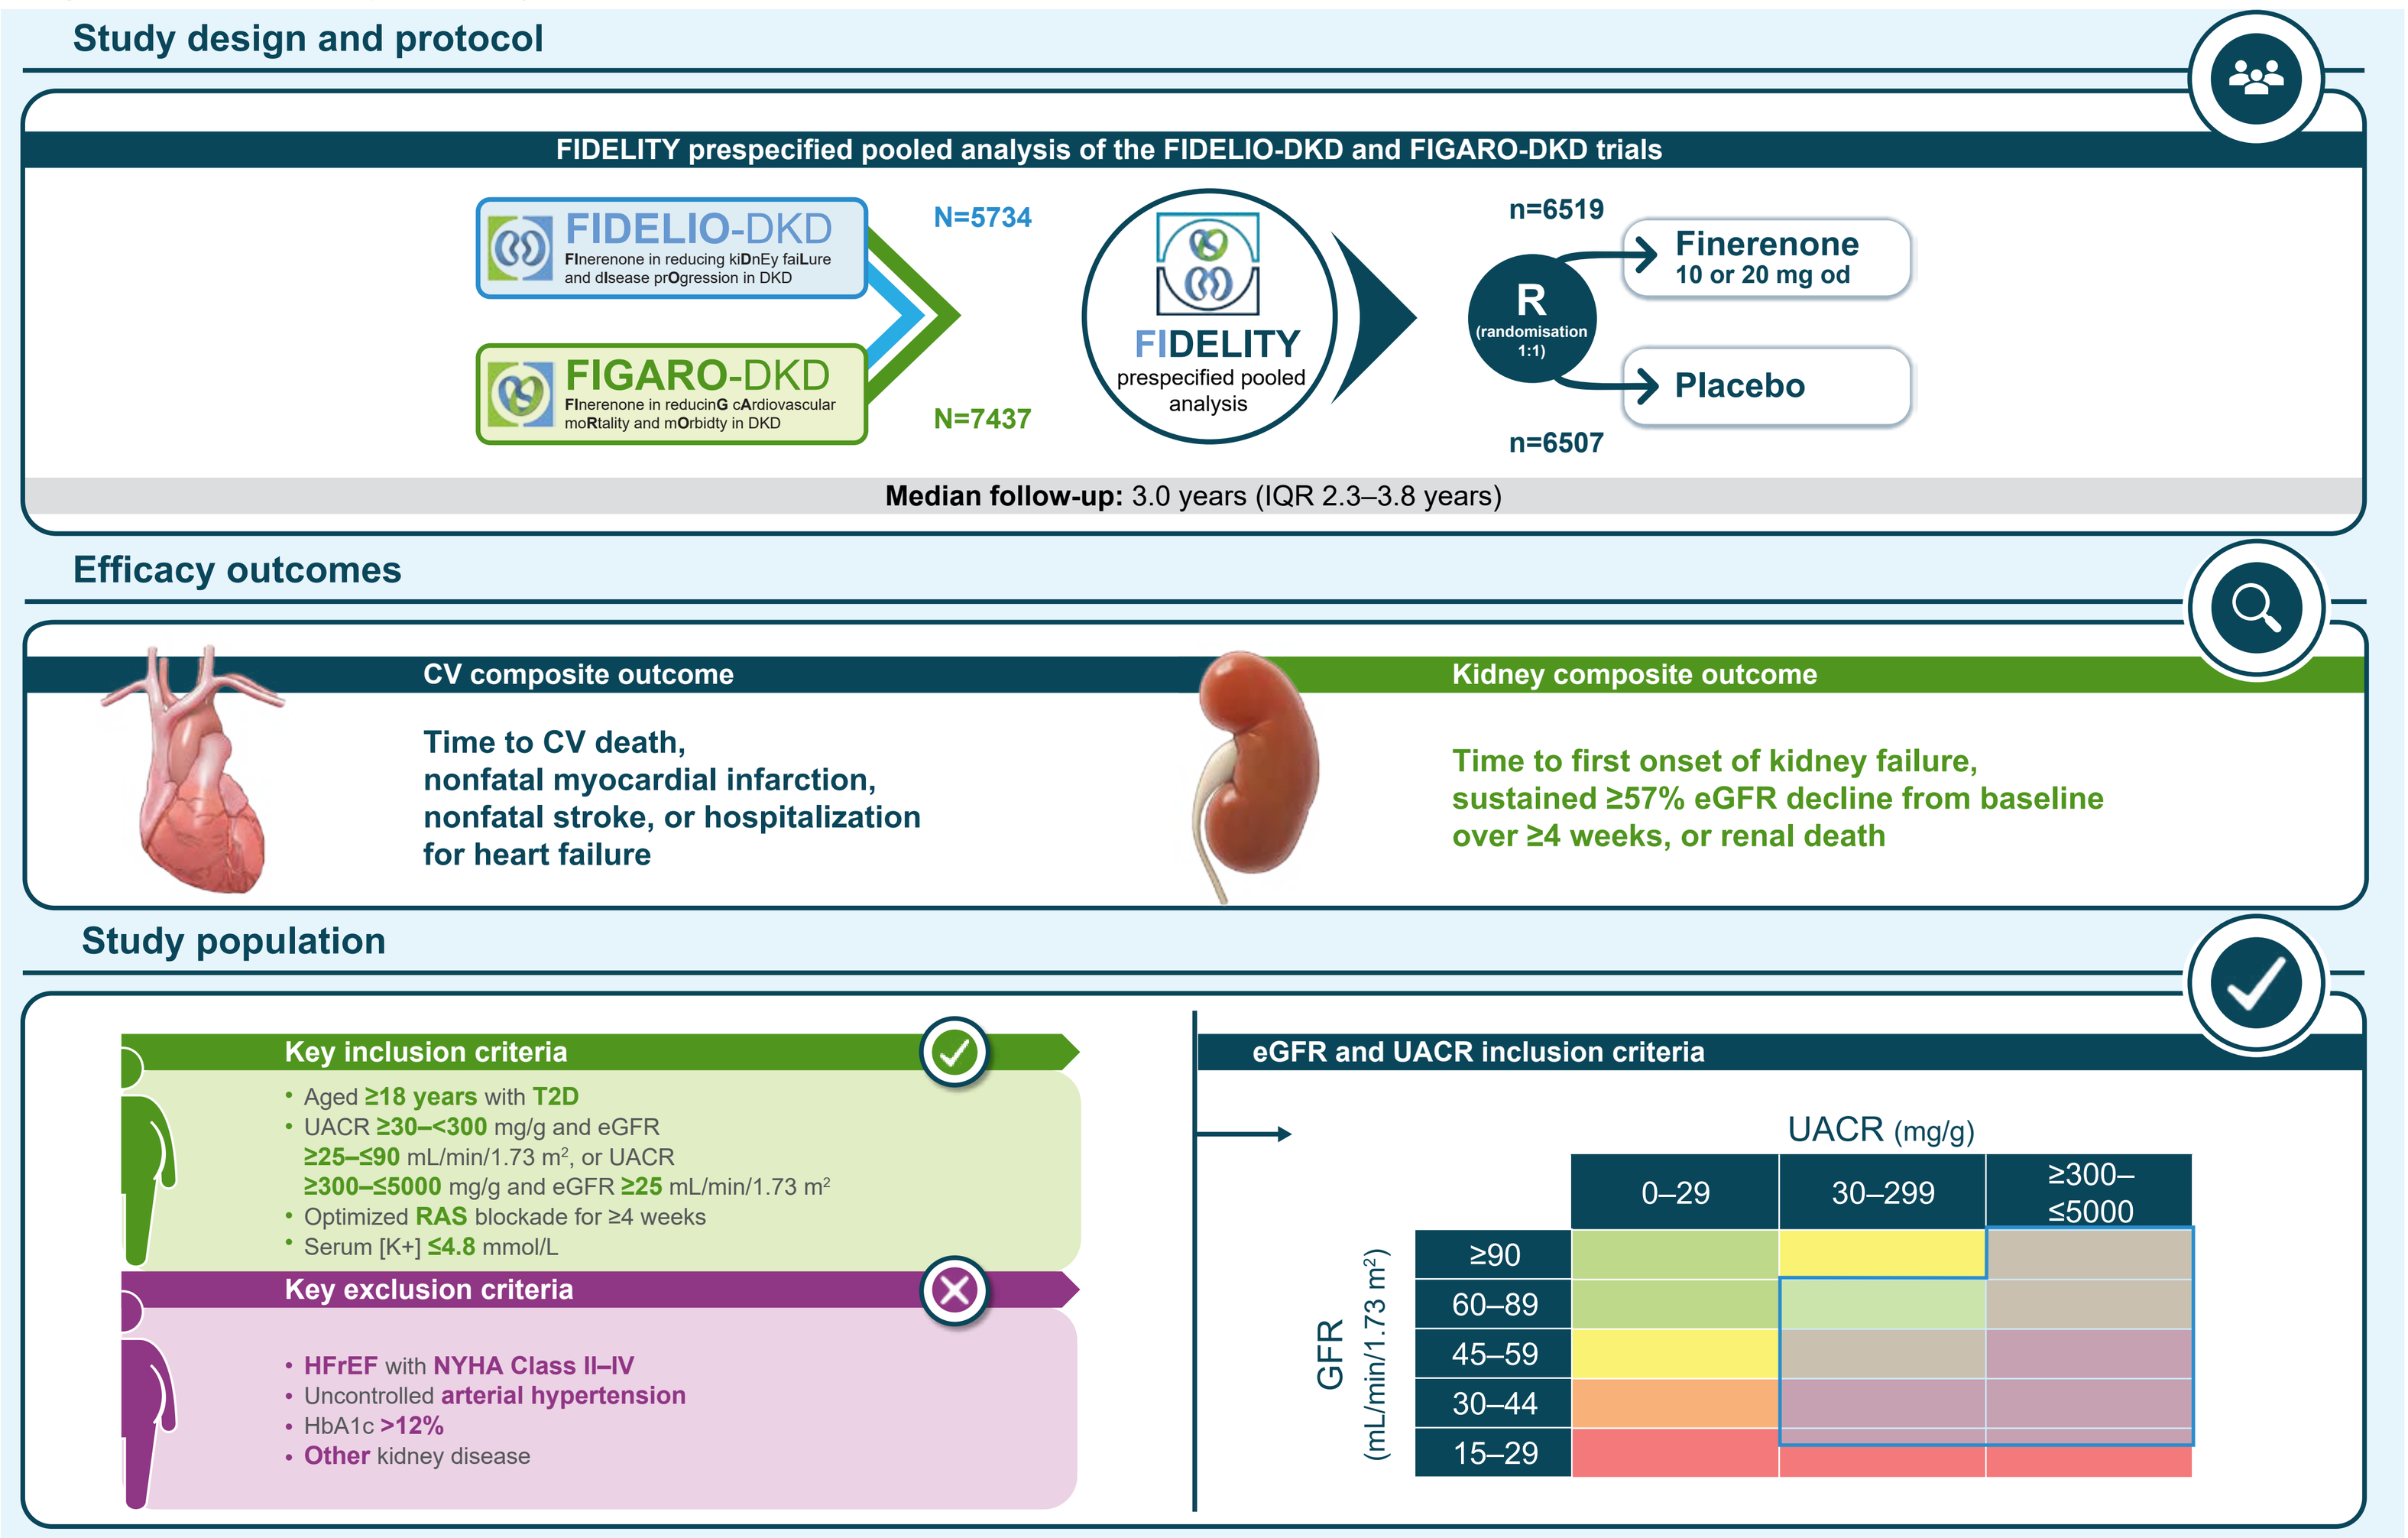
Study design and protocol
FIDELITY prespecified pooled analysis of the FIDELIO-DKD and FIGARO-DKD trials
FIDELIO-DKD
FInerenone in reducing kiDnEy faiLure and dIsease prOgression in DKD
N=5734
FIGARO-DKD
FInerenone in reducinG cArdiovascular moRtality and mOrbidty in DKD
N=7437
FIDELITY
prespecified pooled analysis
R
(randomisation 1:1)
n=6519
Finerenone
10 or 20 mg od
Placebo
n=6507
Median follow-up: 3.0 years (IQR 2.3–3.8 years)
Efficacy outcomes
CV composite outcome
Time to CV death, nonfatal myocardial infarction, nonfatal stroke, or hospitalization for heart failure
Kidney composite outcome
Time to first onset of kidney failure, sustained ≥57% eGFR decline from baseline over ≥4 weeks, or renal death
Study population
Key inclusion criteria
Aged ≥18 years with T2D
UACR ≥30–<300 mg/g and eGFR ≥25–≤90 mL/min/1.73 m², or UACR ≥300–≤5000 mg/g and eGFR ≥25 mL/min/1.73 m²
Optimized RAS blockade for ≥4 weeks
Serum [K+] ≤4.8 mmol/L
Key exclusion criteria
HFrEF with NYHA Class II–IV
Uncontrolled arterial hypertension
HbA1c >12%
Other kidney disease
eGFR and UACR inclusion criteria
UACR (mg/g)
0–29
30–299
≥300–≤5000
GFR (mL/min/1.73 m²)
≥90
60–89
45–59
30–44
15–29

CV, cardiovascular; DKD, diabetic kidney disease; eGFR, estimated glomerular filtration rate; GFR, glomerular filtration rate; HbA1c, glycated hemoglobin; HFrEF, heart failure with reduced ejection fraction; IQR, interquartile range; [K+], potassium concentration; NYHA, New York Heart Association; od, once daily; RAS, renin–angiotensin system; T2D, type 2 diabetes; UACR, urine albumin-to-creatinine ratio

3. Results

- Among 12,964 patients included in the analysis, 6485 (50%) patients received finerenone treatment and 6479 (50%) received placebo
 - Median baseline eGDR was 4.1 mg/kg/min; 6484 (50%) patients had an eGDR <median (with insulin resistance) and 6480 (50%) had an eGDR ≥median (without insulin resistance) (**Table 1**)
 - Overall, baseline characteristics were well balanced between groups, with some notable differences. Patients with an eGDR <median had a longer mean duration of diabetes, and higher median urine albumin-to-creatinine ratio and mean weight, compared with patients with an eGDR ≥median.

Table 1. Patient baseline characteristics according to insulin resistance at baseline

Baseline Characteristic	eGDR at baseline			
	eGDR <median		eGDR ≥median	
	Finerenone (n=3247)	Placebo (n=3237)	Finerenone (n=3238)	Placebo (n=3242)
Age, years, mean	64.5	64.6	64.9	65
Sex, female, n (%)	926 (28.5)	897 (27.7)	1096 (33.8)	992 (30.6)
Duration of diabetes, years, mean ± SD	16.2 ± 8.6	16.1 ± 8.5	14.7 ± 8.8	15.0 ± 8.8
HbA1c, %, mean ± SD	8.2 ± 1.4	8.2 ± 1.4	7.2 ± 1.1	7.2 ± 1.1
BMI, kg/m², mean ± SD	34.6 ± 5.7	34.6 ± 5.6	28.1 ± 4.4	28.0 ± 4.3
Weight, kg, mean ± SD	99.1 ± 18.7	99.3 ± 18.7	76.9 ± 14.8	77.0 ± 14.1
SBP, mmHg, mean ± SD	138.5 ± 14.0	138.1 ± 13.9	135.1 ± 14.1	135.3 ± 14.5
History of CV disease, n (%)	1565 (48.2)	1615 (49.9)	1396 (43.1)	1331 (41.1)
eGFR, mL/min/1.73 m², mean ± SD	57.7 ± 21.9	57.5 ± 21.9	57.4 ± 21.3	57.8 ± 21.6
UACR, mg/g, median	529.7	542.8	494	492
Serum potassium, mmol/L, mean ± SD	4.4 ± 0.4	4.4 ± 0.4	4.4 ± 0.4	4.4 ± 0.5
Baseline medications, n (%)				
ACE inhibitors	1483 (45.7)	1516 (46.8)	1290 (39.8)	1315 (40.6)
ARBs	2015 (62.1)	2045 (63.2)	2173 (67.1)	2179 (67.2)
Beta blockers	2226 (68.6)	2241 (69.2)	1584 (48.9)	1665 (51.4)
Diuretics	2446 (75.3)	2495 (77.1)	1809 (55.9)	1873 (57.8)
Statins	2672 (82.3)	2681 (82.8)	2448 (75.6)	2498 (77.1)
Potassium supplements	337 (10.4)	376 (11.6)	230 (7.1)	289 (8.9)
Potassium-lowering agents	245 (7.5)	142 (4.4)	281 (8.7)	197 (6.1)
Glucose-lowering therapies, n (%)				
Insulin and analogs	2298 (70.8)	2229 (68.9)	1551 (47.9)	1519 (46.9)
Sulfonylureas	769 (23.7)	760 (23.5)	914 (28.2)	933 (28.8)
DPP-4 inhibitors	704 (21.7)	698 (21.6)	951 (29.4)	909 (28.0)
GLP-1RAs	356 (11.0)	300 (9.3)	137 (4.2)	144 (4.4)
SGLT-2 inhibitors	275 (8.5)	268 (8.3)	162 (5.0)	170 (5.2)

ACE, angiotensin-converting enzyme; ARB, angiotensin receptor blocker; BMI, body mass index; CV, cardiovascular; DPP-4; dipeptidyl peptidase-4; eGDR, estimated glucose disposal rate; eGFR, estimated glomerular filtration rate; GLP-1RA; glucagon-like peptide-1 receptor agonist; HbA1c, glycated hemoglobin; SBP, systolic blood pressure; SD, standard deviation; SGLT-2, sodium-glucose co-transporter-2; UACR, urine albumin-to-creatinine ratio

3.1. Efficacy outcomes

- Analysis by continuous baseline eGDR showed a significantly lower risk of CV events at 3.5 years with increasing eGDR (hazard ratio [HR]=0.88 [95% confidence interval (CI) 0.86–0.91]; *p*<0.01), whereas baseline eGDR was not associated with the incidence of kidney outcomes (**Figure 2**)
 - Similar trends were observed when considering baseline eGDR subgroups, where eGDR <median was associated with a higher incidence rate (IR) of the CV composite outcome versus eGDR ≥median (IR per 100 patient-years 5.18 and 3.47 in the finerenone group and 6.34 vs 3.76 in the placebo group, respectively); the IR of the kidney composite outcome was similar across eGDR subgroups (**Figure 3**)
- There was no significant heterogeneity for the effect of finerenone by baseline eGDR on the CV outcomes or kidney outcomes (**Figure 3**)
- Consistent strength and direction of the associations were observed across sensitivity analyses using alternative measures of insulin resistance (baseline triglyceride/high-density lipoprotein ratio, visceral adiposity index, and lipid accumulation product index)

Figure 2. CV and kidney composite outcomes by continuous variable eGDR

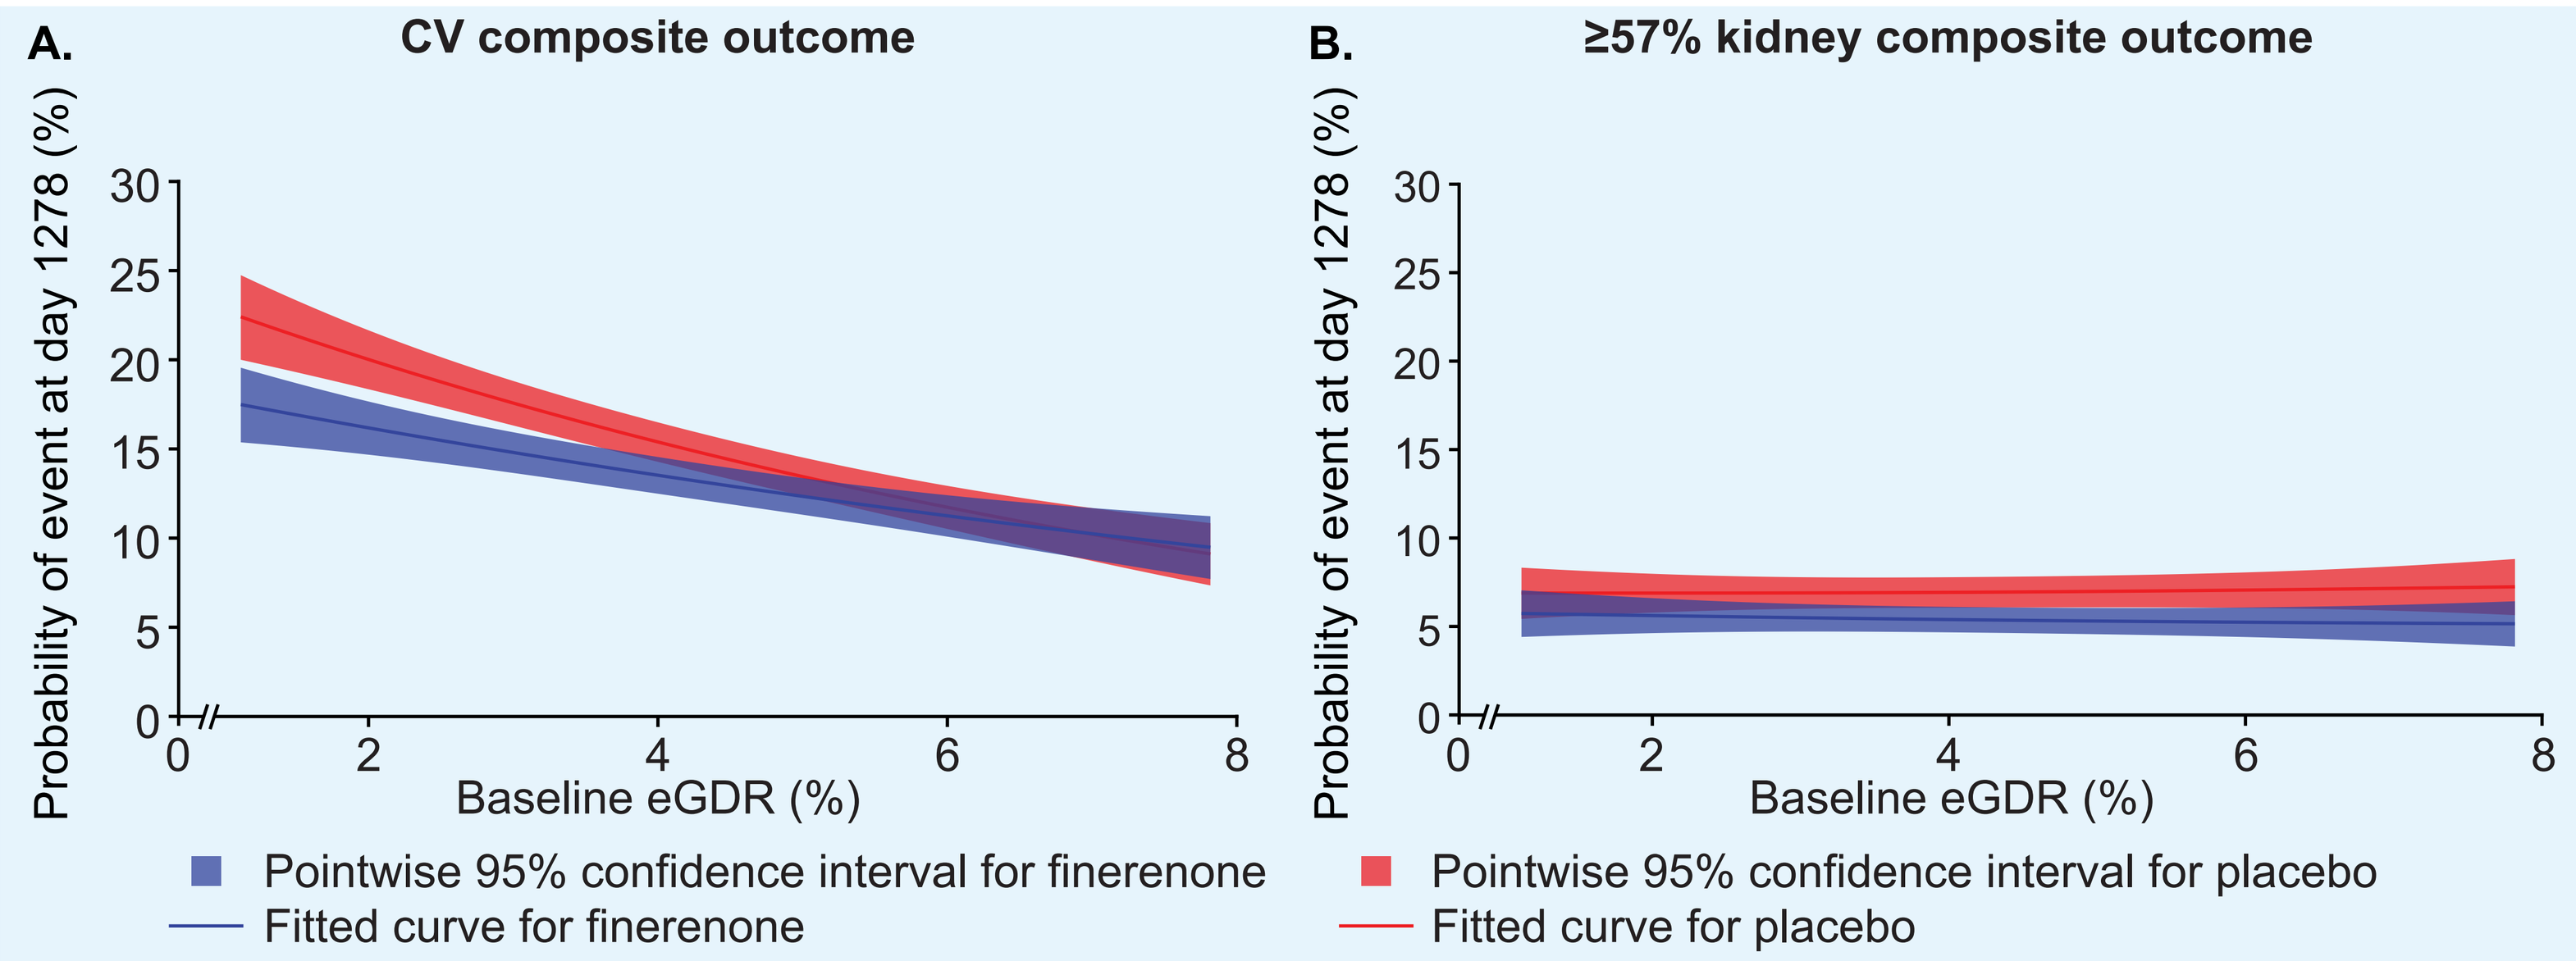
A.
CV composite outcome
B.
≥57% kidney composite outcome
Probability of event at day 1278 (%)
0
5
10
15
20
25
30
0
2
4
6
8
Baseline eGDR (%)
Pointwise 95% confidence interval for finerenone
Fitted curve for finerenone
Pointwise 95% confidence interval for placebo
Fitted curve for placebo

CV, cardiovascular; eGDR, estimated glucose disposal rate

Figure 3. CV and kidney outcomes by baseline eGDR

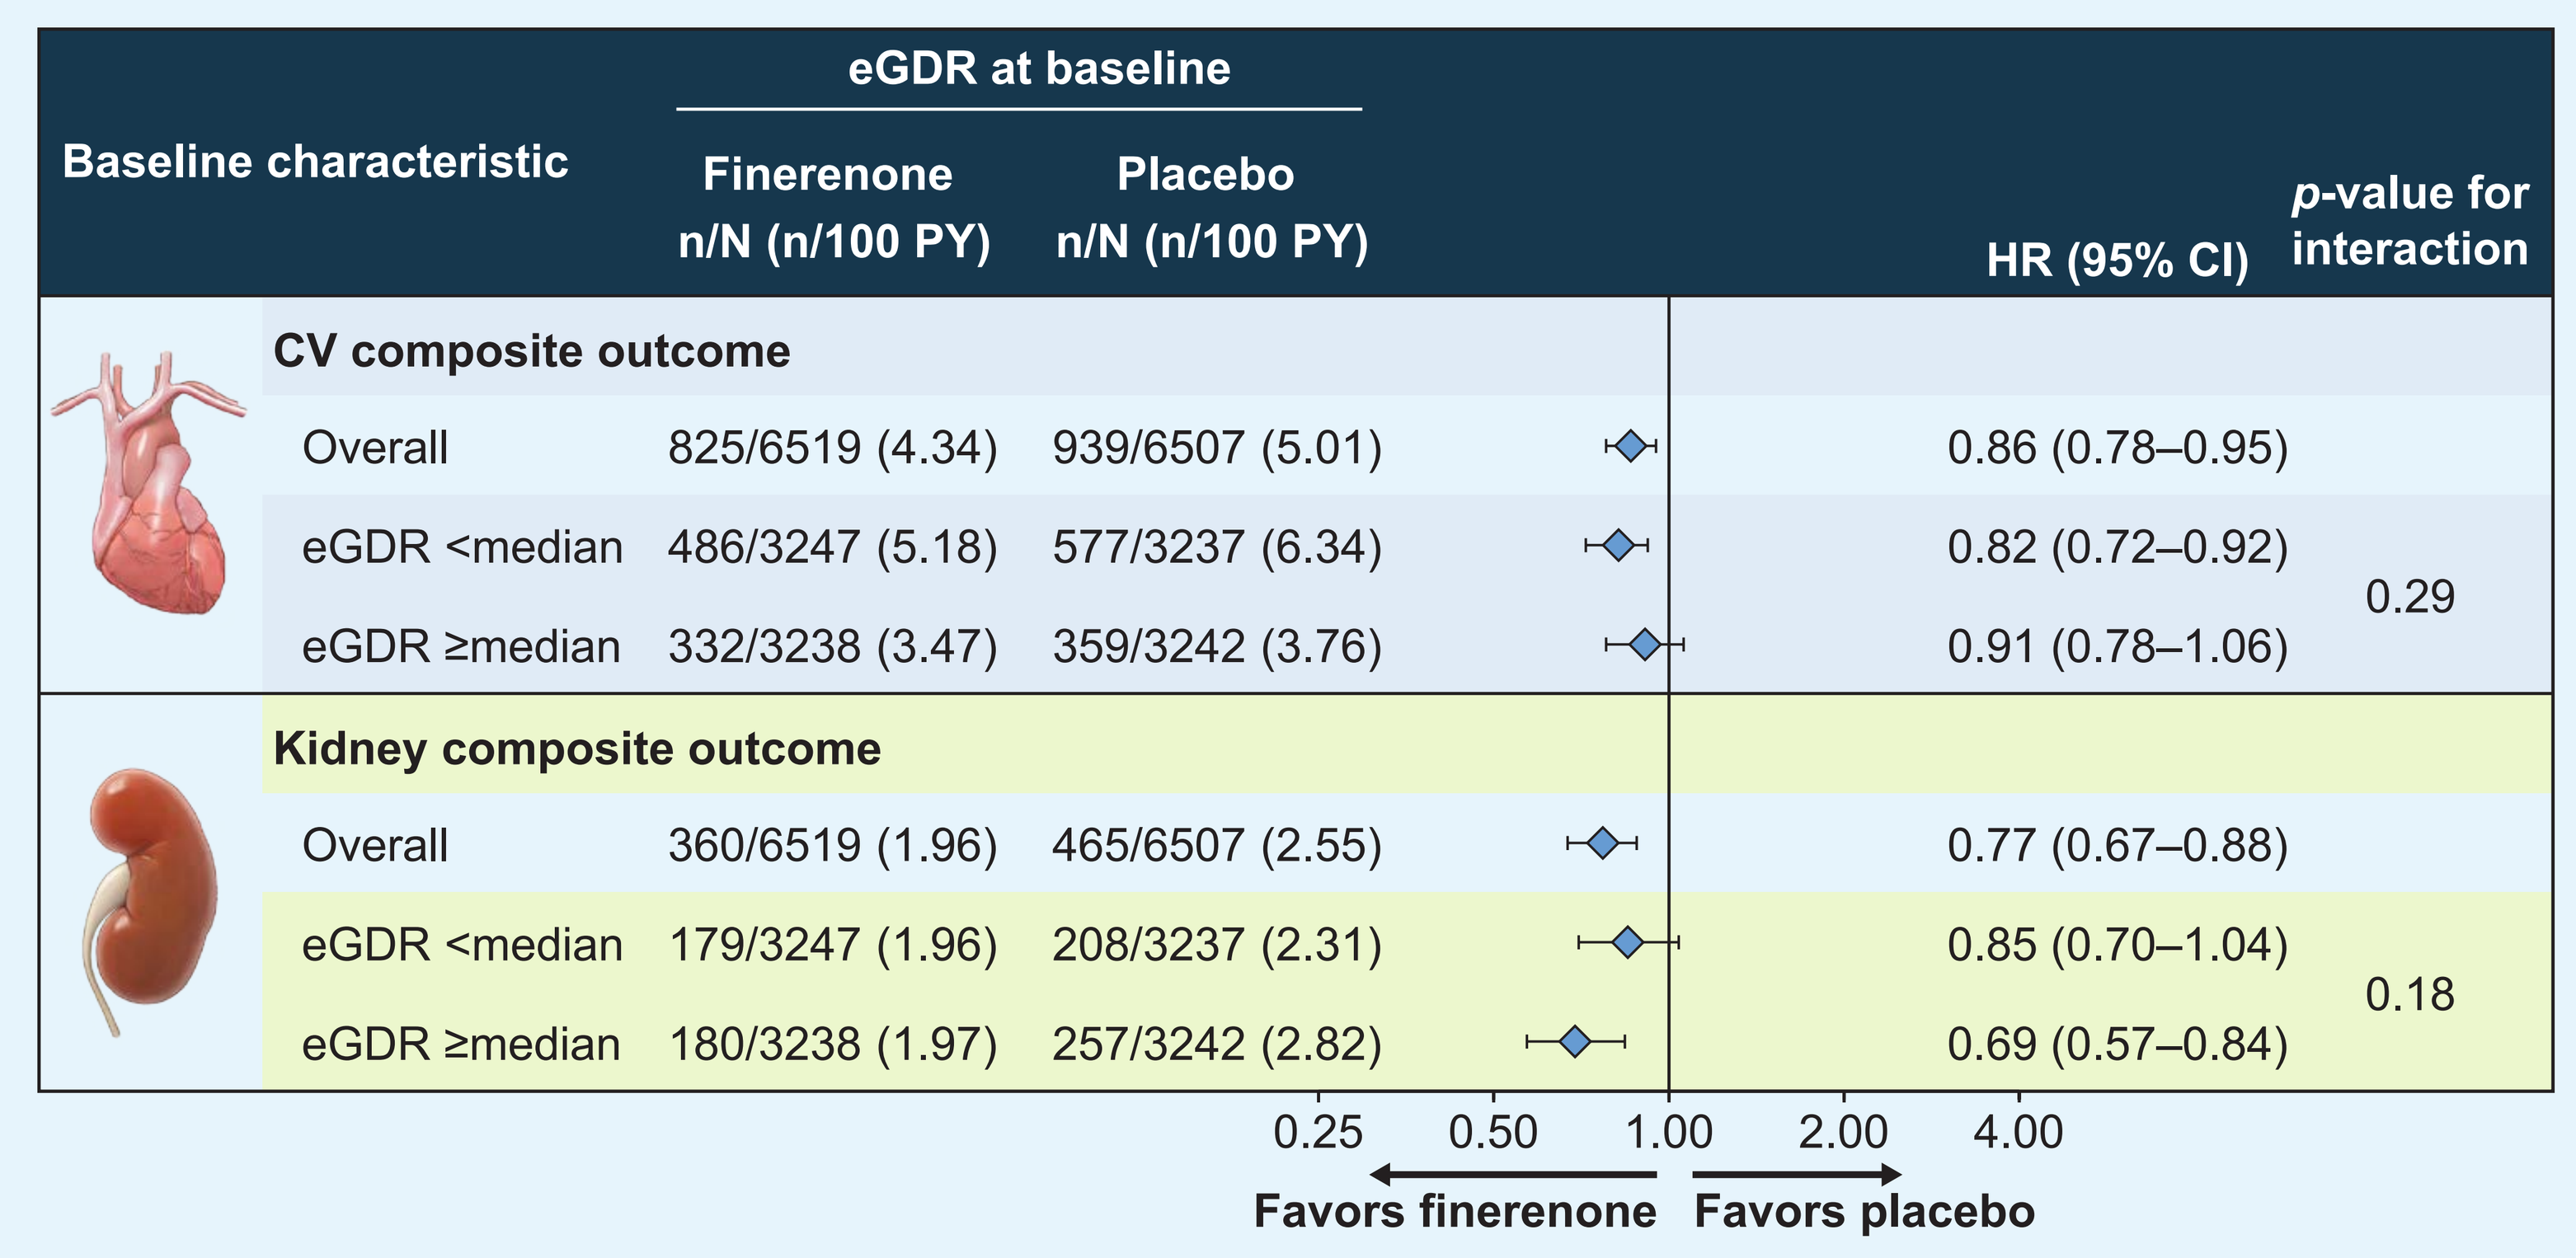

Baseline characteristic	eGDR at baseline: Finerenone n/N (n/100 PY)	eGDR at baseline: Placebo n/N (n/100 PY)	HR (95% CI)	*p*-value for interaction
CV composite outcome				
Overall	825/6519 (4.34)	939/6507 (5.01)	0.86 (0.78–0.95)	
eGDR <median	486/3247 (5.18)	577/3237 (6.34)	0.82 (0.72–0.92)	0.29
eGDR ≥median	332/3238 (3.47)	359/3242 (3.76)	0.91 (0.78–1.06)	
Kidney composite outcome				
Overall	360/6519 (1.96)	465/6507 (2.55)	0.77 (0.67–0.88)	
eGDR <median	179/3247 (1.96)	208/3237 (2.31)	0.85 (0.70–1.04)	0.18
eGDR ≥median	180/3238 (1.97)	257/3242 (2.82)	0.69 (0.57–0.84)	

0.25
0.50
1.00
2.00
4.00
Favors finerenone
Favors placebo

CI, confidence interval; CV, cardiovascular; eGDR, estimated glucose disposal rate; HR, hazard ratio; PY, patient-years

3.1. Safety outcomes

- Overall, the incidences of treatment-emergent adverse events and severe adverse events were balanced between the finerenone and placebo groups and between eGDR subgroups (**Table 2**)
 - The incidence of investigator-reported, treatment-emergent hyperkalemia was higher in patients treated with finerenone versus placebo in both eGDR subgroups. However, hyperkalemia leading to discontinuation was low in the finerenone treatment group, with no notable differences between eGDR subgroups (eGDR <median: 1.9%; eGDR ≥median: 1.5%)

Table 2. Safety outcomes according to insulin resistance at baseline (safety analysis set)

Treatment-emergent adverse events, n (%)	eGDR at baseline			
	eGDR <median		eGDR ≥median	
	Finerenone (n=3242)	Placebo (n=3228)	Finerenone (n=3235)	Placebo (n=3234)
Treatment-emergent adverse events				
Any AE	2823 (87.1)	2801 (86.8)	2751 (85.0)	2781 (86.0)
Study drug-related AE	640 (19.7)	457 (14.2)	560 (17.3)	402 (12.4)
AE leading to discontinuation	236 (7.3)	170 (5.3)	176 (5.4)	180 (5.6)
Any SAE	1107 (34.1)	1181 (36.6)	937 (29.0)	999 (30.9)
Study drug-related SAE	46 (1.4)	32 (1.0)	36 (1.1)	29 (0.9)
SAE leading to discontinuation	84 (2.6)	72 (2.2)	59 (1.8)	82 (2.5)
Fatal AE	55 (1.7)	83 (2.6)	54 (1.7)	68 (2.1)
Treatment-emergent hyperkalemia events				
Any AE	460 (14.2)	195 (6.0)	449 (13.9)	252 (7.8)
Study drug-related AE	286 (8.8)	107 (3.3)	285 (8.8)	142 (4.4)
AE leading to discontinuation	63 (1.9)	19 (0.6)	47 (1.5)	19 (0.6)
Any SAE	36 (1.1)	11 (0.3)	32 (1.0)	5 (0.2)
Study drug-related SAE	22 (0.7)	6 (0.2)	20 (0.6)	2 (<0.1)
SAE leading to discontinuation	8 (0.2)	1 (<0.1)	2 (<0.1)	1 (<0.1)
Fatal AE	0	0	0	0

AE, adverse event; SAE, serious adverse event

4. Conclusions

- In this post hoc analysis of the FIDELITY prespecified pooled analysis, the efficacy and safety of finerenone were not modified by baseline insulin resistance
- A higher risk of CV outcomes, but not kidney outcomes, was observed in people with T2D and CKD with greater insulin resistance
- The safety profile of finerenone was generally consistent irrespective of baseline insulin resistance

Acknowledgments

Funded by Bayer AG; FIDELIO-DKD and FIGARO-DKD clinicaltrials.gov number NCT02540993 and NCT02545049, respectively. Medical writing assistance was provided by Chameleon Communications International and was funded by Bayer AG.

Disclosures

TE reports personal fees from Bayer, Fresenius Medical Care Deutschland, Sanofi, and Santis. He has received research support from the European Foundation for the Study of Diabetes (EFSD Mentorship Programme supported by AstraZeneca). **SA** has received research support from Abbott Vascular and Vifor International, and personal fees from Abbott Vascular, Bayer, Boehringer Ingelheim, BRAHMS, Cardiac Dimensions, Impulse Dynamics, Novartis, Servier, and Vifor Pharma. **LR** reports receipt of consultancy fees from Bayer. **PF** has served as an advisory board member and speaker for AstraZeneca, Eli-Lilly, Bayer, Novo Nordisk, and Mundipharma. **VF** has served as a paid consultant for Abbott, Asahi, AstraZeneca, Bayer, Novo Nordisk, and Sanofi. He has patent and ownership interests in BRAVO4Health. **GU** has received research support (to Emory University) from AstraZeneca, Bayer, Abbott, and Dexcom Inc. **AB** reports personal fees from AstraZeneca, Boehringer Ingelheim, and Novo Nordisk during the conduct of the study; all fees are given to the University Clinic Tübingen. **RL**, **CS**, and **KR** are full-time employees of Bayer. **PR** reports personal fees from Bayer during the conduct of the study. He has received research support and personal fees from AstraZeneca and Novo Nordisk, and personal fees from Astellas Pharma, Boehringer Ingelheim, Eli Lilly, Gilead, Mundipharma, Sanofi, and Vifor Pharma; all fees are given to Steno Diabetes Center Copenhagen.

References

1. Whaley-Connell A, Sowers JR. *Cardiorenal Med* 2017;8:41–49.
2. Schrauben SJ, et al. *BMC Nephrol* 2019;20:60.
3. Nakashima A, et al. *Nutrients* 2021;13.
4. Agarwal R, et al. *Eur Heart J* 2022;43:474–484.
5. Bakris GL, et al. *N Engl J Med* 2020;383:2219–2229.
6. Pitt B, et al. *N Engl J Med* 2021;385:2252–2263.

Poster 29-P presented at the American Diabetes Association (ADA) 2023 (June 23–26, 2023, San Diego, CA, USA)